Hepdart 2021: Abstract P12

LIMT-2: A PHASE 3 STUDY OF 48-WEEK TREATMENT WITH PEGINTERFERON LAMBDA IN PATIENTS WITH CHRONIC HEPATITIS DELTA VIRUS (HDV) INFECTION

Ingrid Choong[1], Nicole Ramza[1], Elizabeth Cooney[1], Colin Hislop[1], Ohad Etzion[2,3] for the *LIMT-2* Clinical Study Investigators

[1]Eiger BioPharmaceuticals, Inc; [2]Faculty of Health Sciences, Ben-Gurion University of the Negev, Israel; [3]Department of Gastroenterology and Liver Diseases, Soroka University Medical Center, Israel

PURPOSE

To evaluate the safety and efficacy of Peginterferon Lambda in a global, registrational study of 150 patients with chronic HDV.

BACKGROUND

Hepatitis Delta Virus (HDV) infection leads to the most severe form of human viral hepatitis for which there is no FDA-approved therapy. HDV is always a coinfection with HBV and is found in approximately 6% of HBV infected patients. Worldwide prevalence of HDV infection is 15-20 million. However, HDV causes a much more rapid progression of liver disease than HBV alone. 60% of HDV patients die within 10 years after diagnosis. This is a large orphan disease in the U.S. and Europe with an urgent, unmet medical need.

Peginterferon Lambda is a well-characterized, first-in-class, type III interferon (IFN) that stimulates immune responses that are critical for the development of host protection during viral infections. Peginterferon Lambda targets type III IFN receptors which are distinct from the type I IFN receptors targeted by IFN alfa, but results in activation of the same Jak-STAT signal transduction cascade. Peginterferon Lambda type III receptors are highly expressed on hepatocytes with limited expression on hematopoietic and central nervous system cells, which reduces off-target effects and improves tolerability of Peginterferon Lambda.

Peginterferon Lambda is well-characterized with a large safety database of > 3000 HBV, HCV, HDV and COVID-19 patients. In a previous Phase 2 head-to-head study in chronic HBV patients, there were fewer and less severe cases of cytopenia, flu-like and psychiatric symptoms compared to peginterferon alfa.

In a prior Phase 2 study, 33 patients with chronic HDV infection were treated with once-weekly subcutaneous injections of Peginterferon Lambda for 48 weeks. 36% of patients achieved a durable virologic response (DVR) defined as HDV RNA level below the limit of quantitation (BLQ) at 24-weeks post-treatment.

Herein, we describe the Phase 3 *LIMT-2* study, a single, global, pivotal trial of Peginterferon Lambda for the treatment of HDV. *LIMT-2* represents a potentially efficient pathway for Peginterferon Lambda approval in HDV, and is now undergoing site activation and patient screening.

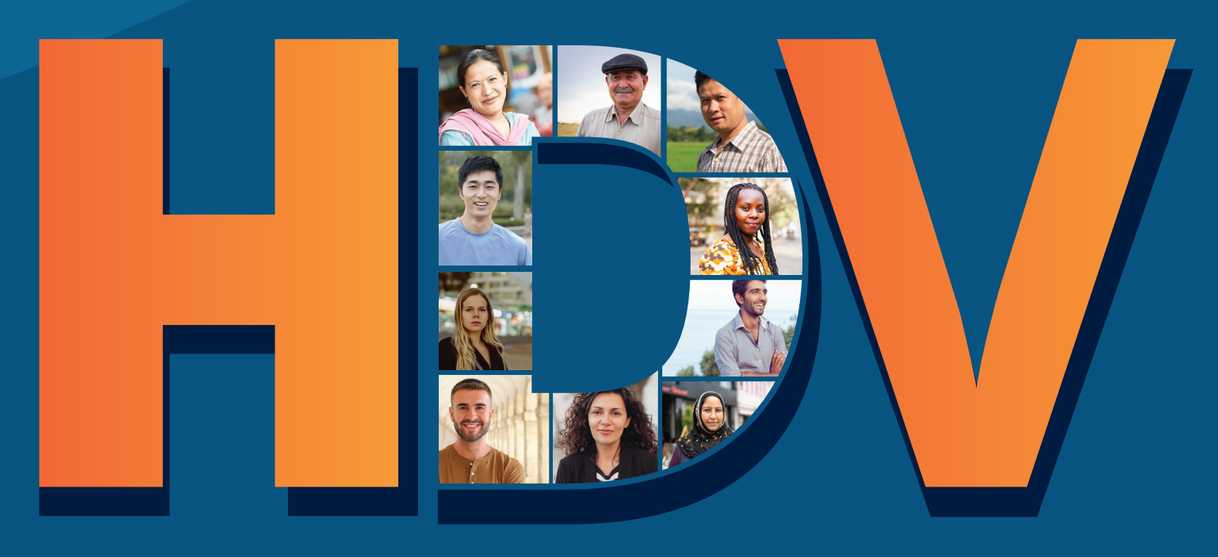
HDV

Peginterferon Lambda for HDV

A WELL TOLERATED INTERFERON

- Binds to a unique receptor vs type I IFN-alfa
 - Highly expressed on hepatocytes
 - Limited expression on hematopoietic and CNS cells
- Uses similar downstream signaling pathway to IFN-alfa
- 3,000+ patients in 19 clinical trials (HCV / HBV / HDV / COVID-19)
- **Orphan Designation** in U.S. and EU
- FDA **Breakthrough Therapy Designation**
- Composition of matter and method of use patents

Lambda Receptors Highly Expressed in the Liver

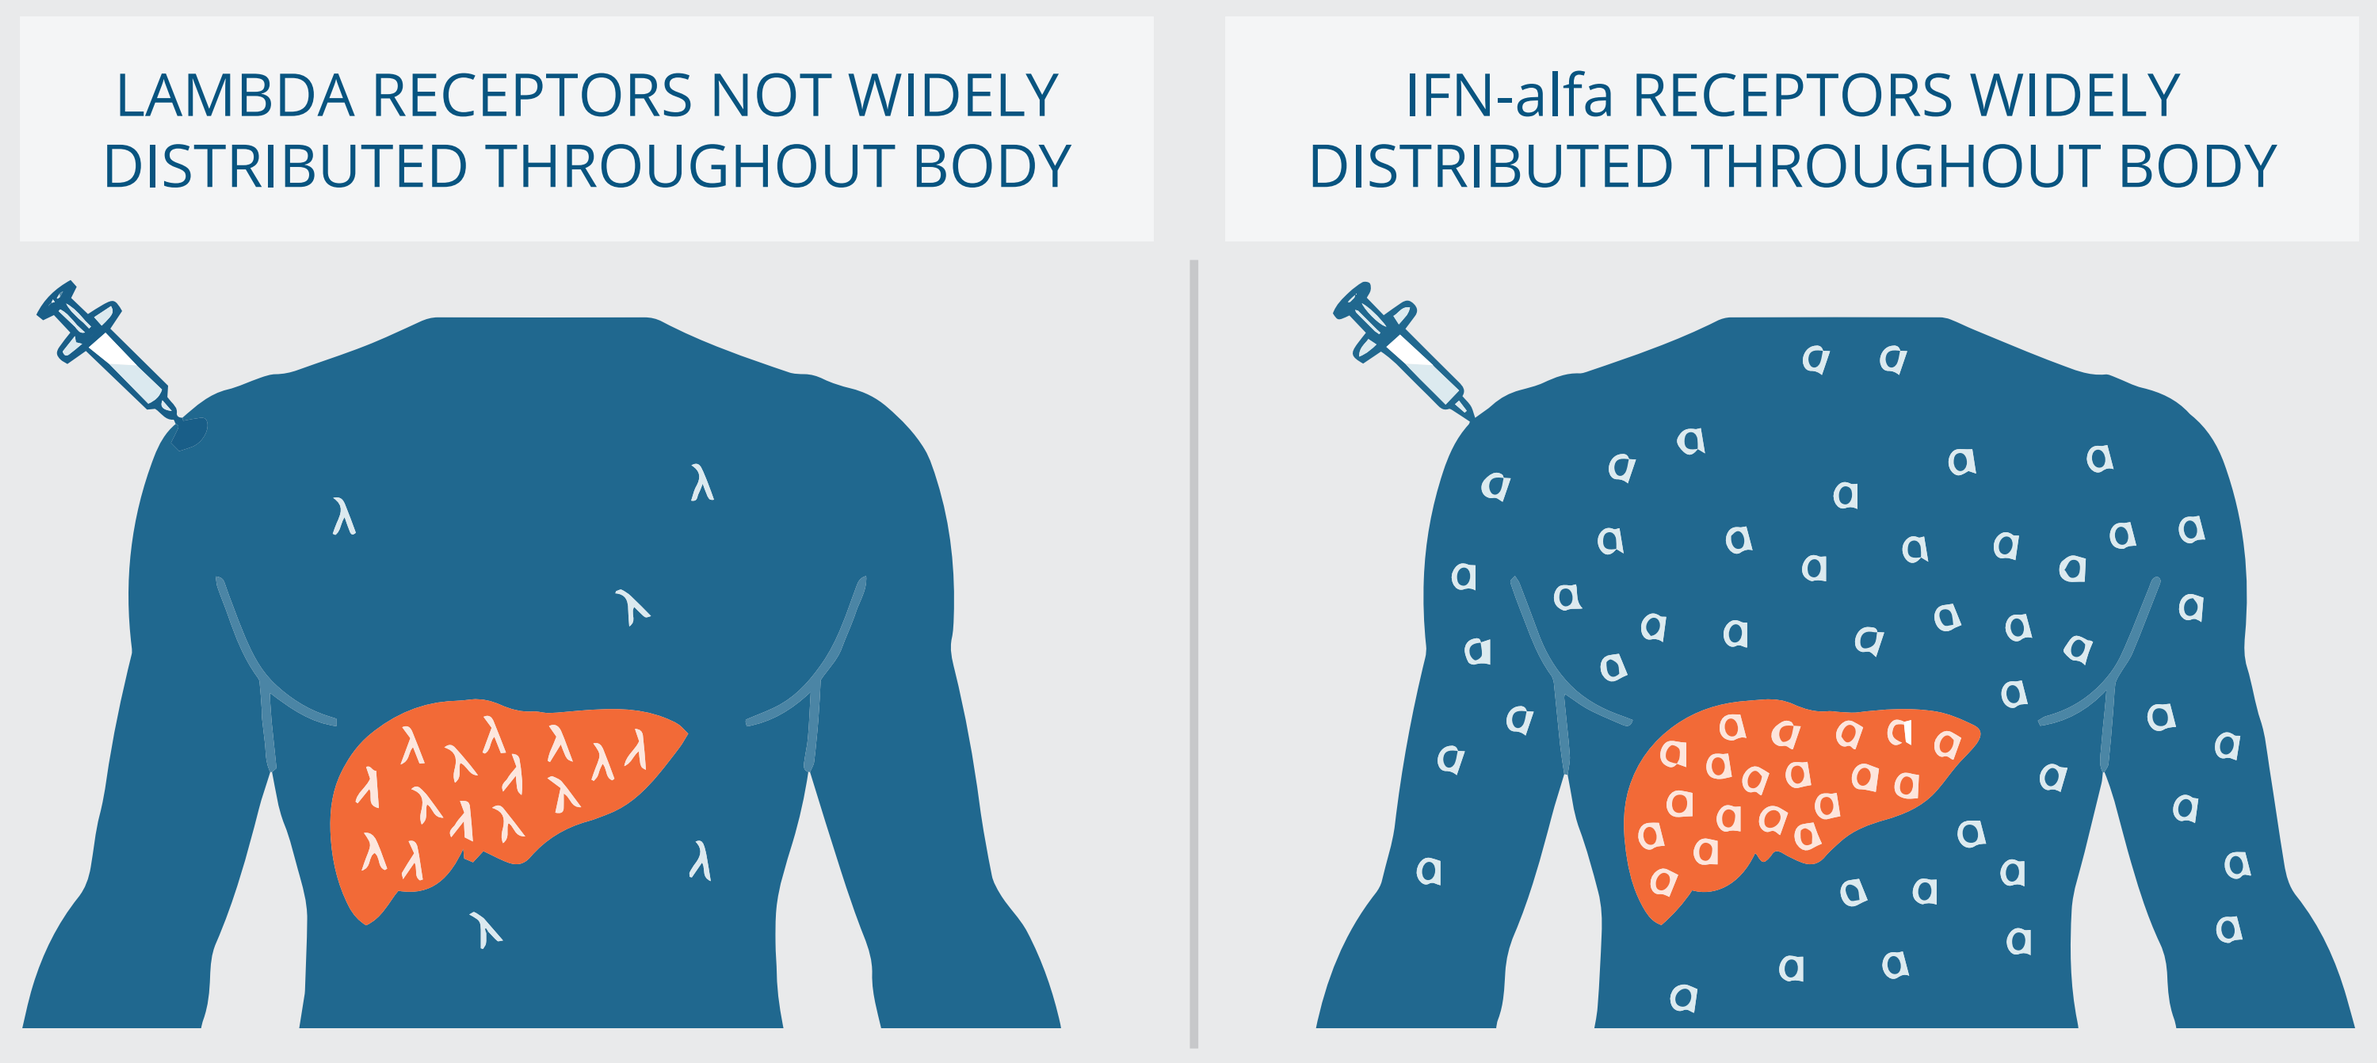
LAMBDA RECEPTORS NOT WIDELY DISTRIBUTED THROUGHOUT BODY
IFN-alfa RECEPTORS WIDELY DISTRIBUTED THROUGHOUT BODY

METHODS

Design

Phase 3, multicenter, randomized, open-label study in patients with chronic HDV infection

Treatment

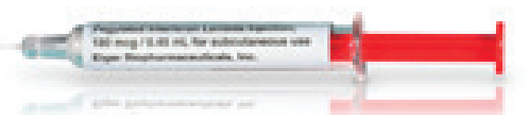

Peginterferon Lambda is a pre-filled syringe that will be self-administered once weekly for 48 weeks

Study Sites

50 sites across 13 countries

Key Features of *LIMT-2*

- Patients who complete 12 weeks of no treatment in Arm 2 will be eligible to receive Peginterferon Lambda for 48 weeks

Phase 3 *LIMT-2* Study Design

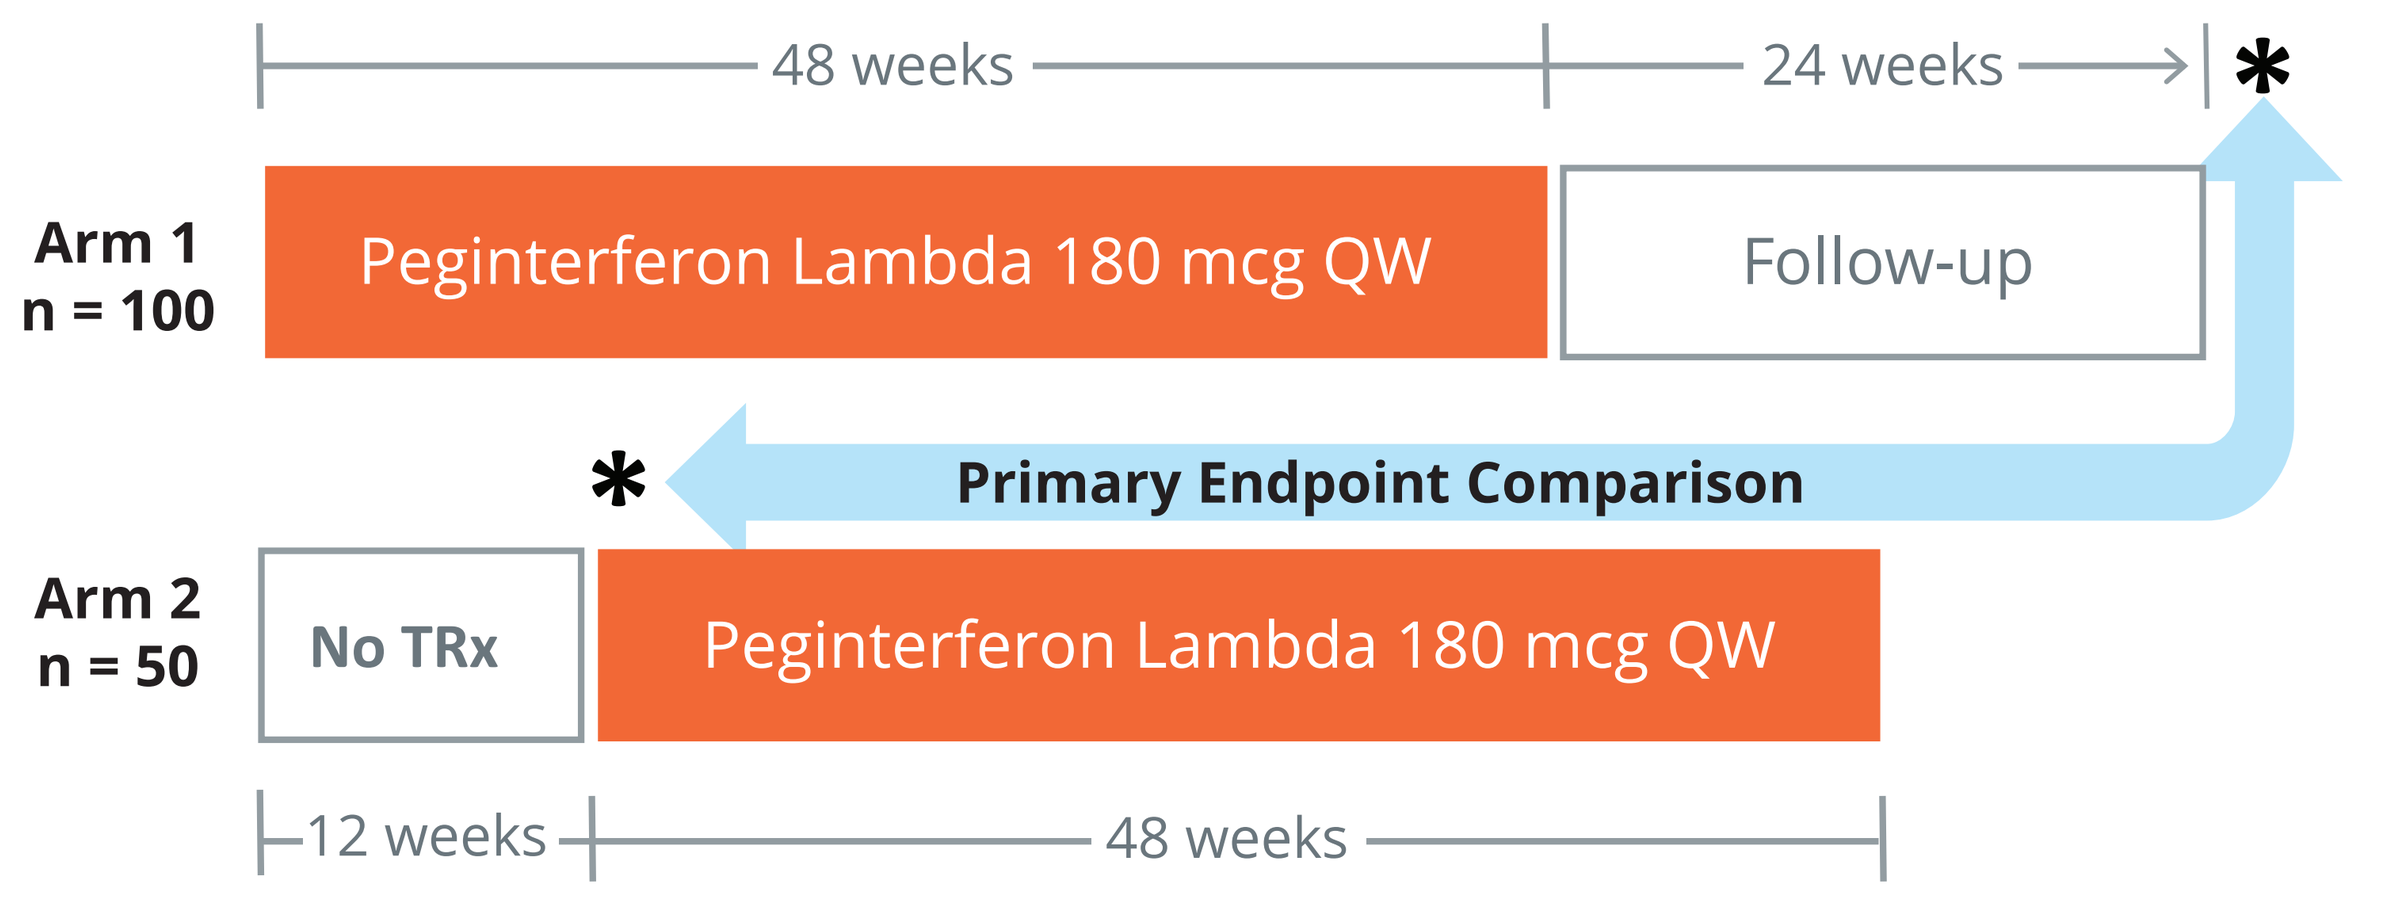
48 weeks
24 weeks
*
Arm 1
n = 100
Peginterferon Lambda 180 mcg QW
Follow-up
*
Primary Endpoint Comparison
Arm 2
n = 50
No TRx
Peginterferon Lambda 180 mcg QW
12 weeks
48 weeks

All patients will receive HBV background therapy (tenofovir or entecavir).

***Primary Endpoint:**

DVR (Arm 1) vs HDV RNA BLQ after 12 Weeks of No Treatment (Arm 2)

DVR (Durable Virologic Response) = Below the Limit of Quantification (BLQ) at 24 Weeks Post-Treatment

RESULTS

Phase 3 *LIMT-2* Study Sites: 50 sites across 13 countries

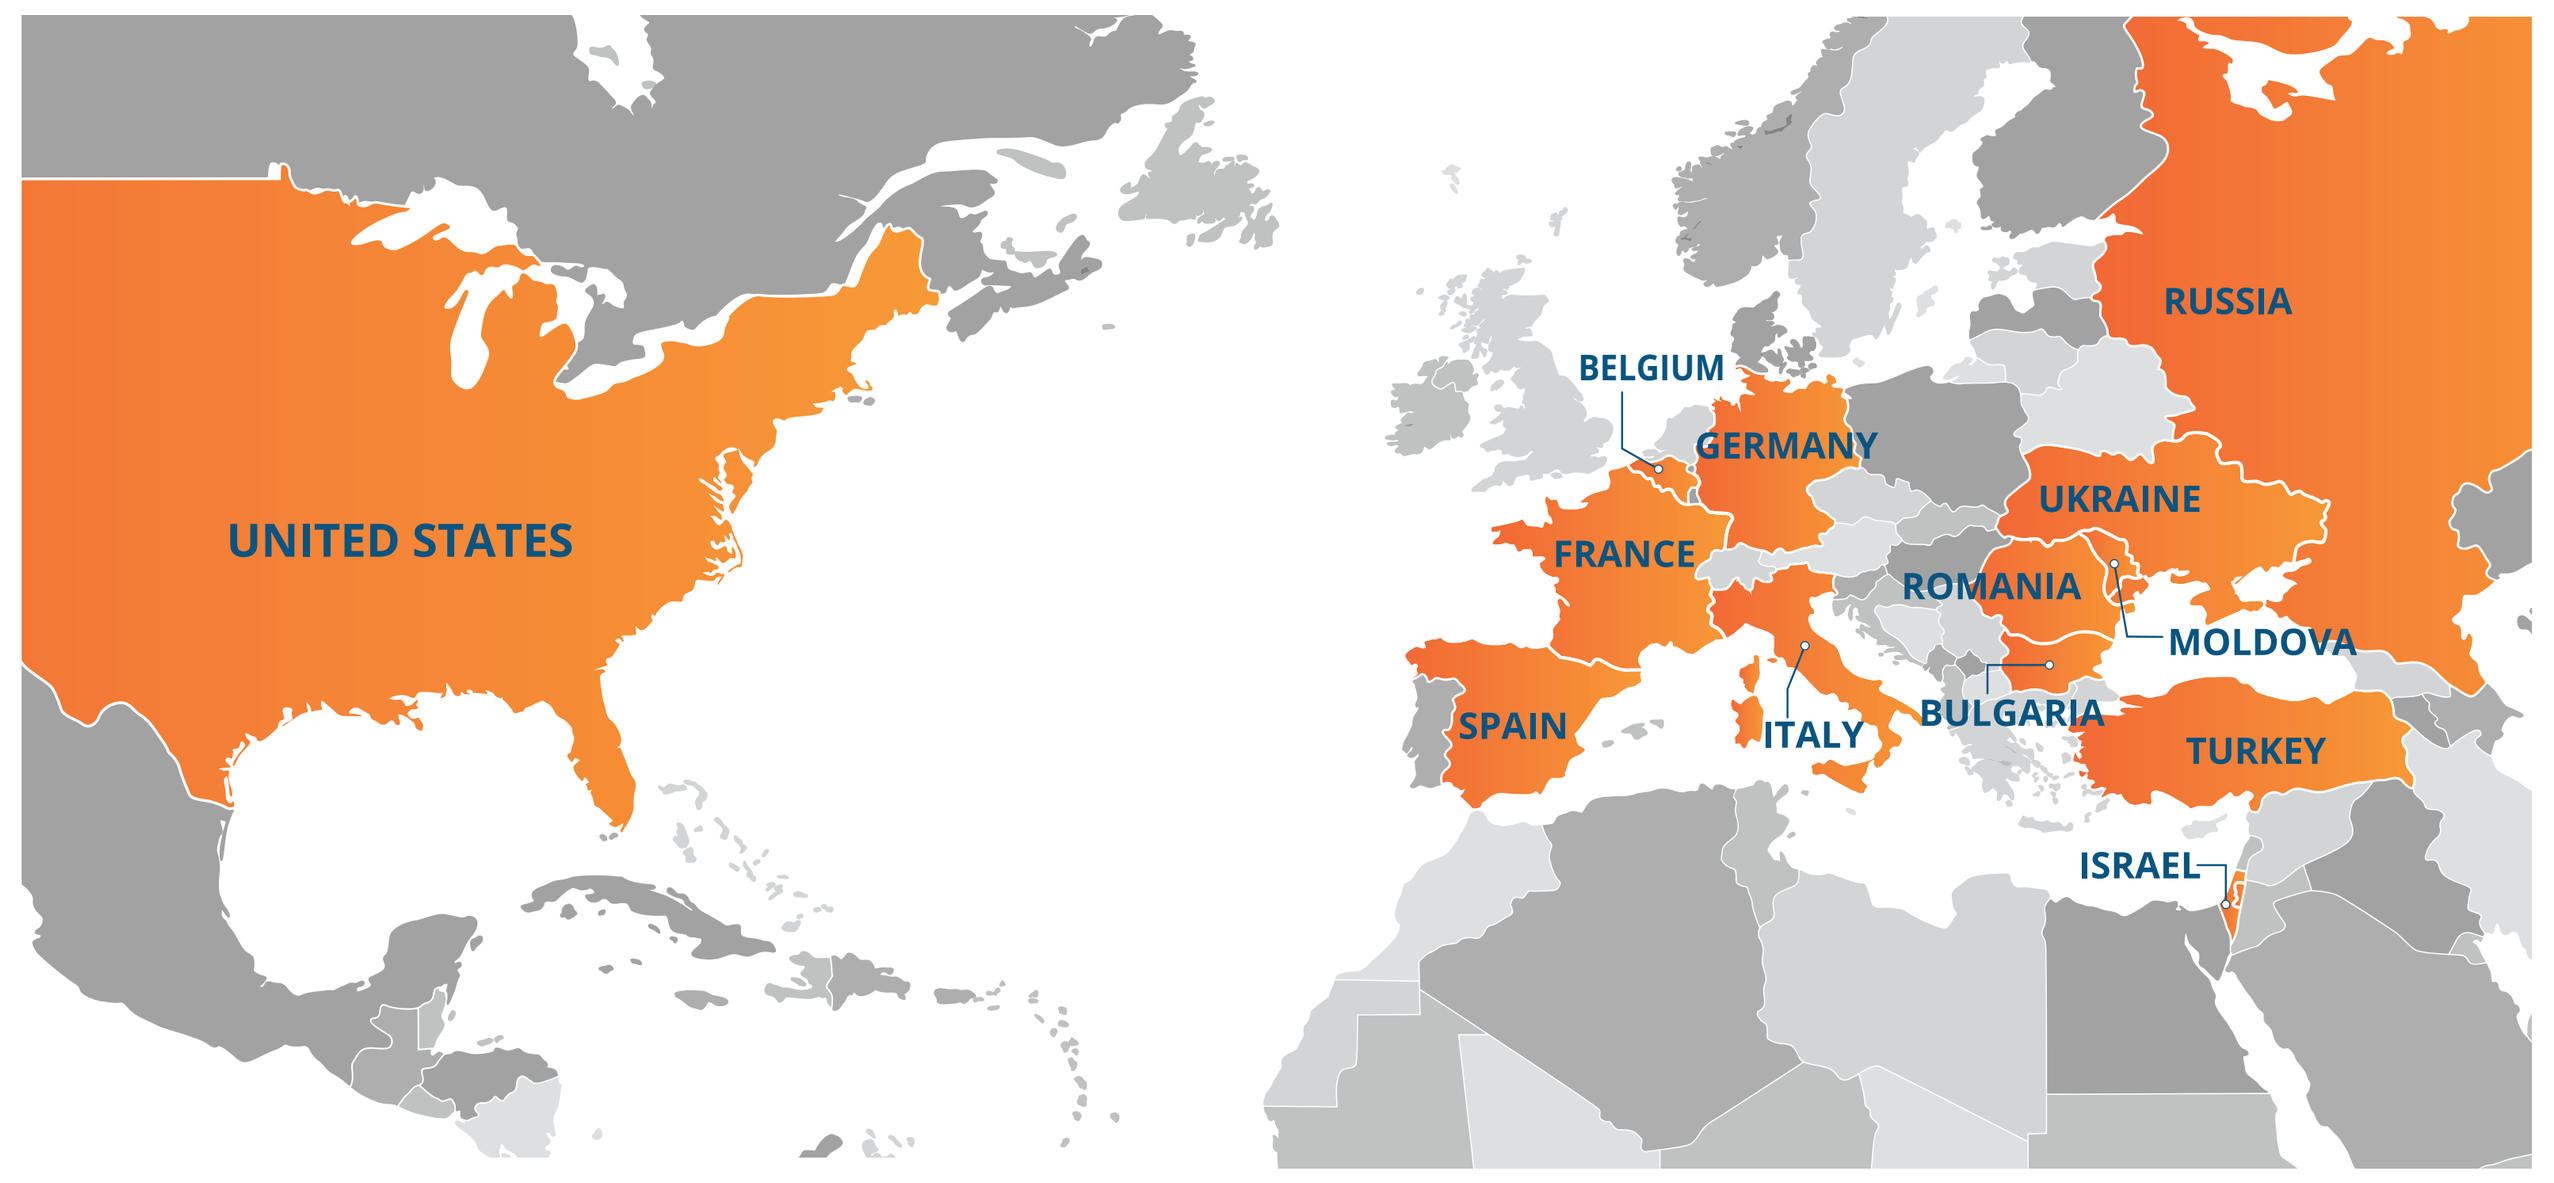
UNITED STATES
BELGIUM
GERMANY
RUSSIA
UKRAINE
FRANCE
ROMANIA
MOLDOVA
SPAIN
ITALY
BULGARIA
TURKEY
ISRAEL

Key Eligibility Criteria

Inclusion Criteria

- Chronic HDV infection, confirmed by > 6 months of infection
- Quantifiable HDV RNA by RT-PCR
- Suppression of HBV DNA (< 100 IU/mL) following at least 12 weeks of anti-HBV NUC treatment
- Serum ALT > upper limit of normal (ULN) and < 10 × ULN
- Child-Turcotte-Pugh score of ≤ 5 with well compensated liver disease

Exclusion Criteria

- History or current evidence of decompensated liver disease (episodes of variceal bleeding, ascites or encephalopathy)
- Treatment with interferons (IFNs) or immunomodulators within 12 months of randomization or refractory response to prior IFN treatment

CONCLUSION

Screening has initiated in ***LIMT-2*** with the first patient expected to be randomized in 2021. The primary analysis will compare the proportion of patients with a DVR, or HDV RNA BLQ at 24-weeks post-treatment in the Peginterferon Lambda treatment group (Arm 1) to the proportion of patients with HDV RNA BLQ after 12 weeks of no treatment in the comparator group (Arm 2). Approximately 150 patients will be enrolled in 13 countries across 50 investigator sites. Enrollment to this study will be competitive.

www.clinicaltrials.gov: NCT05070364

Country/City	Institution	Principal Investigator
Belgium		
Brussels	CHU Brugmann	Luc, L.
Brussels	CUB Hôpital Erasme	Moreno, C.
Edegem	University Hospital Antwerp	Vanwolleghem, T.
Ghent	University Hospital Ghent	Degroote, H.
Bulgaria		
Sofia	MC “Nov Rehabilitatsionen Tsentar” EOOD	Penkova, M.
Stara Zagora	Acibadem city clinic Tokuda Hospital	Balabanska, R.
France		
Bobigny	APHP, hôpital Avicenne	Ganne, N.
Clichy	Hôpital Beaujon, Clichy	Asselah, T.
Creteil	Henri-Mondor University Hospital	Ingiliz, P.
Toulouse	Chu Toulouse	Metivier, S.
Germany		
Frankfurt	University Hospital Frankfurt	Sprinzl, K.
Hannover	Medizinische Hochschule Hannover	Wedemeyer, H.
Mainz	Universitätsmedizin Mainz	Grambihler, A.
Israel		
Beer-Sheva	Soroka University Medical Center	Etzion, O.
Haifa	Liver Disease Center, Rambam Health Care Campus	Veitsman, E.
Jerusalem	Hadassah University Hospital	Safadi, R.
Ramat-Gan	Sheba Medical Center	Ben-Ari, Z.
Italy		
Foggia	Azienda Ospedaliero Universitaria Ospedali Riuniti di Foggia	Santantonio, T.
Milan	University of Milan	Lampertico, P.
Pisa	Pisa University	Brunetto, M.
San Giovanni Rotondo	Casa Sollievo della Sofferenza	Niro Grazia, A.
Moldova		
Chisinau	Gastroenterology and Hepatology Clinic	Turcanu, A.
Romania		
Bucharest	Dr Victor Babes Foundation	Gherlan, G.
Bucharest	Fundeni Clinical Institute	Iliescu, E.
Bucharest	National Institute for Infectious Diseases “Matei Bals”	Carantu, F.
Constanta	Spitalul Clinic de Boli Infectioase Constanta	Dumitry, I.
Galati	Spitalul de Infectioase Galati Romania	Baroiu, L.
Russia		
Krasnodar	Krasnodar Specialized Clinical Infectious Diseases Hospital	Bakhtina, V.
Moscow	Center Tagetnoy Terapii, LLC	Bogomolov, P.
Moscow	H-Clinic, LLC	Voronkova, N.
Moscow	Modern Medicine Clinic	Stepanova, T.
Novosibirsk	Medical Center "Healthy Family"	Voloshina, N.
Samara	Hepatolog, LLC	Morozov, V.
Stavropol	Stavropol State Medical University	Geyvandova, N.
Spain		
Alcorcón	Hospital Universitario Fundación Alcorcón	Gutiérrez García, M.
Barcelona	Hospital Universitario Vall d`Hebron	Buti, M.
Turkey		
Istanbul	Istanbul University-Cerrahapasa	Tabak, F.
Istanbul	Koç University Medical School	Yurdaydin, C.
Sur Diyarbakir	Dicle University Hospital	Celen, M.
Ukraine		
Ivano-Frankivsk	Ivano-Frankivsk National Medical University	Oleksandra, P.
Kyiv	Medical Center of LLC “Harmoniya Krasy”	Anastasii, I.
Kyiv	Medical Center ‘Ok!Clinic+’ of International Institute of Clinical Studies LLC	Tsarynna, N.
Sumy	Sumy State University Clinic	Chemych, M.
Ternopil	Ternopil University Hospital	Hosppdarskyy, I.
Vinnytsia	Communal Noncommercial Enterprise Vinnytsia City Clinical Hospital #1	Vasylivna, M.
USA		
Chicago, IL	Rush University Medical Center	Reau, N.
Los Angeles, CA	Asian Pacific Liver Center	Bae, H.
Los Angeles, CA	University of Southern California	Terrault, N.
New York, NY	Mount Sinai Hospital	Dieterich, D.
San Francisco, CA	California Pacific Medical Center	Cooper, S.